THE PATHOGENESIS OF VASCULAR CALCIFICATION, NEW CLINICAL DIAGNOSTIC MARKERS AND A NEW CURATIVE NANOBIOTIC TREATMENT FOR REVERSING ATHEROSCLEROSIS IN HUMANS

Kajander EO[1,2,3], Aho KM[1], Maniscalco BS[3], Mezo GS[3],

[1]University of Kuopio, Kuopio, [2]Nanobac Oy, Kuopio, Finland;

[3]NanobacLabs Research Institute, Tampa, USA

Background:

Atherosclerosis is an inflammatory disease stimulated by various infectious agents. Potential contributing agents should be present at least occasionally in blood. Kajander showed over 10 years ago that human blood contains tiny particles (Fig. 1D) that carry calcium phosphate as their coat, or cell wall[1](Fig. 1A) and to which immunoassays could be developed (Fig. 1B). The agent could cause long infection in spite of an antibody response, entered reticuloendothelial cells in rabbit[2] and in rat circulation[3] and was excreted into urine[3], where it was associated with kidney damage[4] and kidney stones[5]. The particle has been shown to fulfil Koch's postulates in causing kidney stones in an animal model[6,7], and actively mediates apatite crystal formation via secreted slime-protein matrix, see Fig. 1C[8]. The agent was classified as nanobacteria (Nanobacterium sanguineum, Ns)[9] to separate it from other biological entities, because Ns have unique and most extraordinary properties[10] not allowing their placement into any established evolutionary branch of life, see Table 1. Early PCR results initially suggested relatedness to bacteria[11], but this was not supported by more reliable tests. Kajander's findings[9] were disputed and claimed as fraud by Finnish fellows[12]. Kajander was freed of all charges but Nature never published any remittal. Recently Price et al[13-16] and Heiss et al[17] have verified that blood indeed contains calcium phosphate carrying particles, and Price suggested them as a cause for atherosclerosis.

Our hypothesis is that Ns, a new self-replicating infectious calcifying agent found in human blood, which causes life-long infections and induces apoptosis, inflammation and immune attack, is causing atherosclerosis in the human vasculature. To verify and further develop the hypothesis, we have collected epidemiological data on nanobacteria markers, established possible mechanisms contributing to atherosclerosis and its complications, and tested clinical efficacy of a new targeted nanobiotic medication to reverse calcified atherosclerotic plaques and microvascular disease in patients with advanced heart disease.

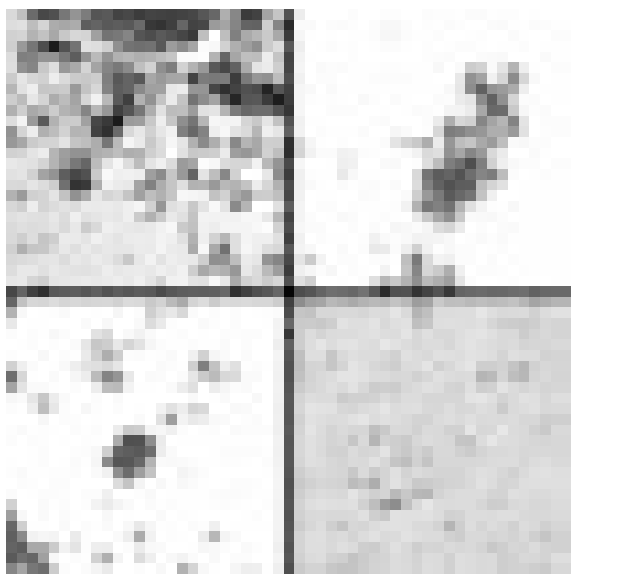

Fig. 1A) TEM reveals Ns structures without any fixation or staining, Bar 1 μm. **(B)** Immunostaining TEM of the same Ns culture using mAb 8D10 conjugated with colloidal gold. Arrow reveals several gold particles adjacent to Ns. Bar 0.12 μm. **(C)** Negative staining using 2% uranyl acetate to reveal mucous protein layer (exampled with arrow) around Ns. This layer is believed to mediate calcification. Bar 1 μm. **(D)** Immunogold staining of uncultured human serum sample, highly positive in Ns test (ELISA). Bar 0.2 μm. Control stainings gave negative results (data not shown).

Table 1. Unique properties of nanobacteria with respect to other prokanyotic-related life-forms.

Property	Nanobacteria	Viral particles	Prion particles	Bacteria
Size	50-300 nm	20-250 nm	<200 nm	>250 nm
Cell wall	CaP/atypical	No/protein layer	No	Yes
Nucleic acids	Some, atypical	Yes, atypical	No	Yes
Proteins	Yes	Yes	Yes	Yes
Carbohydrates	Yes	Yes	Yes	Yes
Self-replicating	Yes	No	No	Yes
Growth in DMEM	Yes	No	No	Yes
Uridine incorporation	Yes	No	No	Yes
Resistant to γ-irradiation	~2.5 Mrad	<2.5 Mrad	>2.5 Mrad	<0.1->6 Mrad
Resistant to boiling temperature	Yes	No	Yes	No
Resists disinfectants	Yes	No/ Yes	Yes	No
Resists antibiotics	No/Yes	Yes	Yes	No/ Yes
Sensitive to 5-FU	Yes	No	No	Yes/No
Sensitive to CytAra	Yes	Yes	No	?
Sensitive to bisphosphonates	Yes	No	No	No
Immunogenic	Yes	Yes	No	Yes
Cause inflammation	Yes	Yes	No	Yes
LPS	Yes	No	No	Yes
Host cell death	Yes	Yes	Specific	Some
Pathologic calcification	Yes	A few	No	A few
Biofilms	Yes	No	No	Yes
CHD association	Yes	Some	No	Some
Stroke association	Yes	Some	No	Some
Affects blood clotting	Yes	Some	No	Some
Prothrombinase activity	Yes	No	No	Some

Methods:

(1) Ns was screened with immunohistochemical staining (Catalyzed Signal Amplification System, DAKO, applied to immunostaining described earlier[18]) in human aortic calcific plaques and with prospective epidemiological survey on Ns markers (Ag and Ab) done by Nyyssönen, Kajander, Ciftcioglu and Salonen. (2) Ns lipid, mineral and protein components and biological interactions were analyzed with focus on atherosclerosis. (3) NanobacTX-ACES Clinical Study was undertaken to document the efficacy of the new nanobiotic, NanobacTX, to reverse coronary artery plaque calcification in 91 patients with well-documented coronary artery disease and high levels of coronary artery calcification. 4-month treatment outcome of NanobacTX was followed with EBCT Coronary Artery Calcification Scoring and with Ns markers in blood. Other medications or diet were not changed during the Study.

Results:

Immunohistochemical staining detected Ns in 2 out of 4 aortic calcific plaques verifying results from the Mayo Clinic[19] and Puskas[20]. Fibrotic cap (see Fig. 2) showed highest Ns antigen positivity followed by deep intimal calcifications and medial aortic wall. The Epidemiological Study found an association between Ns antigen and risk of MI and stroke (Table 2). Apparently Ns marker positivity increased dramatically from 20 to 50 years of age. Ns biomarkers, specifically the antigen showed a very high predictive risk for atherosclerotic complications expected during the 10y follow-up. Ns biofilm contains Ca^{2+}, unique lipopolysaccaride (LPS) and prothrombin, known active components of inflammatory and clotting processes. Specific binding of prothrombin to calcified Ns is shown in Fig 3. Prothrombin has 10 Gla modified amino acids at F1 end which bind to apatite with high affinity. Thrombin cut from prothrombin can be released since it has no Gla residues. Prothrombinase complex contains blood coagulation factors necessary for activation of thrombin, and these factors have Gla residues binding to apatite. Thus, calcified Ns can be loaded with prothrombinase complex and after triggering could release thrombin. Non-calcified Ns did not show any prothrombin on them, see Fig. 3.

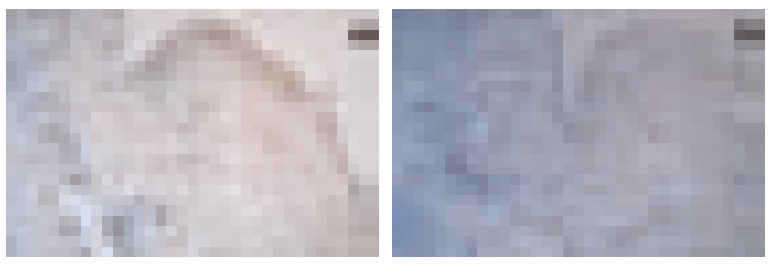

Fig. 2. Fibrous cap area of atherosclerotic plaque stained with anti-Ns mAb 8D10 **(A)** and without mAb **(B)**. CSA staining method was used. Brown color indicated positive Ns antigen. Magnification 105x.

Table 2. Prevalence of Ns markers.

Cohort	Ab positive	Ag positive
~50y initially healthy men, serum sample 1991, 10-year follow-up (n=1033)	90%	32% ***, **, *
~20y students, 2003 (n=58)	24%	5%

***Very high predictive value for MI, **high predictive value for stroke, *statistically significant predictive value for aortic aneurysm incidence during the follow up period.

The NanobacTX-ACES Nanobiotic Study showed decreases in Coronary Artery Calcification EBCT scores by an average of 58% in Study subjects. Fig. 4 shows EBCT calcium scores to regress after 4-months therapy of an individual with a three artery CAD. Normally, this disease is always progressive. Also other risk factors decreased in patients (see Fig 5). In 91 patients participating, there was an average decrease of 58.5% in EBCT scores after 3-month therapy. 19 out of 91 (21%) showed 100% reduction in EBCT scores, i.e. eradication of coronary artery calcification. Ns antigen and antibody showed a trend to rise at 2 months and to fall after 4 months. Preliminary results have been released in Circulation[21].

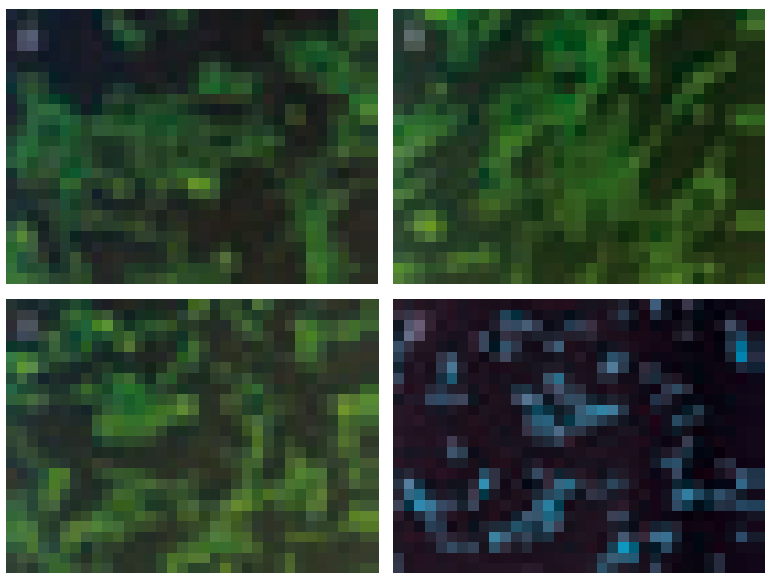

Fig 3. Immunofluorescence staining of BHK cultures exposed to cultured Ns. **(A)**: BHK cells inoculated with standard Ns stained with mAb to human prothrombin (Biodesign, USA) **(B)** BHK cells inoculated with calcium-free Ns, stained as above. **(C)** No inoculation, stained as above. **(D)** Same as **(C)** with Hoechst #33258 to show that no contamination was present in the cultures. Magnification 262x.

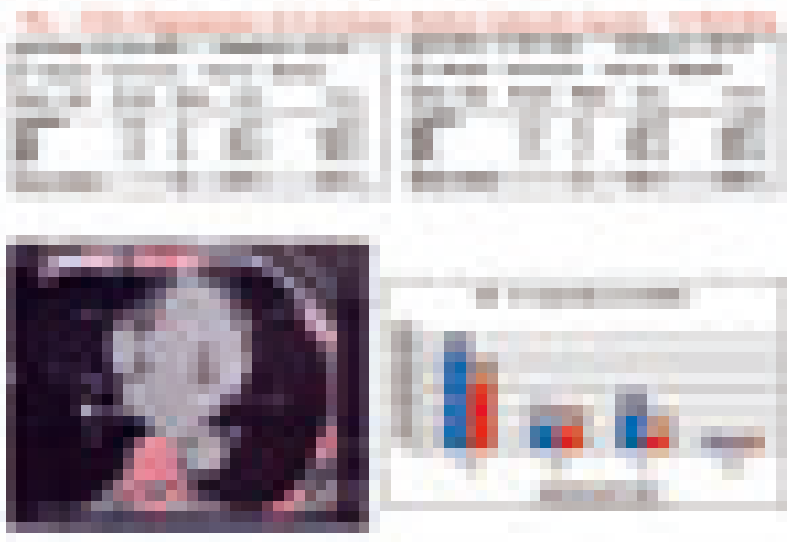

Fig. 4. Representative example of a 23% regression of Coronary artery Calciun Score after 4-month NanobacTX therapy.

Conclusions:

Immunohistochemical staining showed presence of Ns in human aortic calcific plaques. Remarkably, Ns appear to be similar carriers of calcium phosphate than decribed by Price et al[13-16] and Hess et al[17]. Thus Ns could be the calcifiable vesicles isolated earlier from human atherosclerotic aortas[22]. Ns markers in blood have prognostic value for the risk of MI and stroke. Ns contains components capable of stimulating immune cascades leading to local tissue/vascular wall inflammation and are risk factors for arterial and venous thrombus formation. Our earlier animal studies have revealed thrombus formation in large veins and arteries after Ns injection. Nanobiotic treatment with NanobacTX was effective in reversing coronary artery calcification. NanobacTX treatment caused initial increase in Ns markers leading to decline with concomitant improvement of atherosclerotic calcification scores in EBCT. Findings of the study open new insights to pathogenesis, diagnostics and treatment of atherosclerosis. Improvements to our theory on pathogenesis of atherosclerosis[8, 23, 24] is presented in Scheme 1. Further research is clearly warranted to prove the theory.

nanobacTX decreases Cardiac Inflammation & Calcium Scores

Case: Female, 82-CT 53% Decrease at 6 Months & Improved CRP, Lp(a), Fibrinogen

	Baseline	nanobacTX – 4 Months
Fibrinogen	430	400
Lp (a)	45	42
CRP	10.7	3.4
CT Score	2032	1501 - 957 at 6 Months

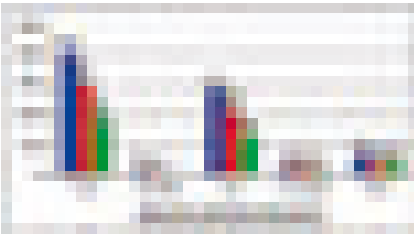

Fig. 5. NanobacTX decreases cardiac inflammation and calcium scores.

MØ

Ns

TOLL-like receptors

Possible co-infection other agents

↓

Persistent infection, cell activation , inflammation, growth factor release, altered endocytic traffic of lipids

↓

FATTY STREAKS

↓

II Middle phase: Increased cell death and inflammation after arousal of immune response, "granulomatous" capping of heavily infected area by fibrous layer

↓

Activation of Ns to form CaP (apatite) intratissue biofilm

↓

CALCIFIC PLAQUE

↓

III Late phase: Revascularization of calcific plaque, spread of highly calcified Ns to fibrous cap

↓

Apoptosis of endothelial cells due to apoptosis induction by Ns, by complement activation, by immunological attack; release of factor V

↓

Exposed Ns layered with CaP bind GLA-proteins (factors II, VII, IX, X) of prothrombinase complex from plasma

↓

Active thrombin (no Gla present) release after contact with platelets

↓

THROMBOSIS

Scheme 1. Theory on atherosclerotic process.

References

1. Kajander EO. Culture and detection method for sterile-filterable autonomously replicating biological particles. US Patent N:o 5,135,851. 16 pp.,1992.
2. Åkerman KK, Kuikka JT, Çiftçioglu N, Parkkinen J, Bergström KA, Kuronen I, Kajander EO. Radiolabeling and in vivo distribution of nanobacteria in rabbit. In: Instruments, Methods, and Missions for the Investigation of Extraterrestrial Microorganisms. Ed. R. B. Hoover, SPIE Proceedings Vol. 3111: 436-442, 1997.
3. Aho K. In vivo biodistribution and toxicity of nanobacteria in rodents. In.: International nanobacteria minisymposium, March 8, 2001, Kuopio, in press.
4. Hjelle JT, Miller-Hjelle MA, Poxton IR, Kajander EO, Çiftçioglu N, Jones ML, Caughey RC, Brown R, Millikin PD, Darras FS. Endotoxin and nanobacteria in polycystic kidney disease. Kidney International 57:2360-2374, 2000.
5. Çiftçioglu N, Björklund M, Kuorikoski K, Bergström K and Kajander EO. Nanobacteria: An infectious cause for kidney stone formation. Kidney International, 56: 1893-1898, 1999.
6. Garcia-Cuerpo E; Kajander EO, Ciftcioglu N, Castellano FL, Correa C, Gonzales J, Mampaso F, Liano F, de Gabiola EG and Berrilero YAE. Nanobacteria. Un modelo de neo-litogenesis experimental. Arch. Esp. Urol. 53(4): 291-303, 2000.
7. Garcia-Cuerpo E, Kajander EO. Litogenesis. Hacia un nuevo planteamiento. Arch. Esp. Urol. 54(9): 851-853, 2001.
8. Kajander EO, Ciftcioglu N, Aho K and Carcia-Cuerpo E. Characteristics of nanobacteria and their possible role in stone formation. Invited Editorial in Urological Research, in press 2003.
9. Kajander EO and Çiftçioglu N. Nanobacteria: An alternative mechanism for pathogenic intra- and extr acellular mineralization and stone formation. Proc Natl Acad Sci USA 95:8274-8279, 1998.
10. Sommer AP, McKay DS, Ciftcioglu N, Oron U, Mester AR, Kajander EO. Living nanovesicles - Chemical and Physical survival strategies of primordial biosystems. J. Proteome Res 1:475, 2002
11. KajanderEO, Kuronen I, Åkerman K , Pelttari A, Çiftçioglu N. Nanobacteria from blood, the smallest culturable autonomously replicating agent on Earth. (An invited article). In: Instruments, Methods, and Missions for the Investigation of Extraterrestrial Microorganisms. Ed. R. B. Hoover, SPIE Proceedings Vol. 3111: 420-428, 1997.
12. Abbott A. Battle lines drawn between 'nanobacteria' researchers. Nature 401(674):105, 1999
13. Price PA, Buckley JR, Williamson MK. The amino bisphosphonate ibandronate prevents vitamin D toxicity and inhibits vitamin D-induced calcification of arteries, cartilage, lungs and kidneys in rats. J Nutr 131:2910-5, 2001
14. Price PA, Faus SA, Williamson MK. Bisphosphonates alendronate and ibandronate inhibit artery calcification at doses comparable to those that inhibit bone resorption. Arterioscler Thromb Vasc Biol 21:817-24, 2001
15. Price PA, Thomas GR, Pardini AW, Figueira WF, Caputo JM, Williamson MK. Discovery of a high molecular weight complex of calcium, phosphate, fetuin, and matrix -carboxyglutamic acid protein in the serum of etidronate-treated rats. J Biol Chem 277:3926-34, 2002
16. Price PA, Omid N, Than TN, Williamson MK. The amino bisphosphonate ibandronate prevents calciphylaxis in the rat at doses that inhibit bone resorption. Calcif Tissue Int.71:356-63, 2002.
17. Heiss A, DuChesne A, Denecke B, Grötzinger J, Yamamoto K, Renne T, Jahnen-Dechent W. Structural basis of calcification inhibition by $_2$-HS Glycoprotein/fetuin-A: formation of colloidal calciprotein particles. J Biol Chem, in press, 2003.
18. Cifcioglu N, Kajander EO Interaction of nanobacteria with cultured mammalian cells. Pathophysiology 4: 259-270, 1998.
19. Rasmunssen et al. JACC 39 Suppl: 206, 2002
20. Puskas, L et al. Detection of nanobacteria in human atherosclerotic plaques. In: International Nanobacteria Minisymposium, March 8, 2001, Kuopio, in press. Abstract can be read at: http://www.nanobac.com/nbminisymp080301/page13.html
21. Maniscalco, B. Shouldering the risk burden: Infection, atherosclerosis, and the vascular endothelium. Circulation, in press March 25, 2003.
22. Hsu HHT, Camacho NP. Isolation of calcifiable vesicles from human atherosclerotic aortas. Atherosclerosis 143: 353-362, 1999.
23. Kajander EO, Ciftcioglu N, Miller-Hjelle MA, Hjelle JT. Nanobacteria: controversial pathogens in nephrolithiasis and polycystic kidney disease. Curr Opin Nephrol Hypertens 10:445-52, 2001
24. Sommer AP, Kajander EO. Nanobacteria-Induced Kidney Stone Formation: Novel Paradigm Based on the FERMIC Model. Crystal Growth & Design, 2:563-565, 2002.